Orally Inhaled Flecainide for the Conversion of Recent-Onset, Symptomatic Atrial Fibrillation to Sinus Rhythm: Results from the INSTANT Phase 2 Study

Harry Crijns[1], Arif Elvan[2], Ype Tuininga[3], Erik Badings[3], Aaf F.M. Kuijper[4], Jonas De Jong[5], Mark Lee[6], Dirk Schellings[7], Isabelle C. Van Gelder[8], A. John Camm[9], Jeremy Ruskin[10], Peter Kowey[11], Prashanti Madhavapeddi[12], Jean Maupas[12], Christopher Dufton[12], Luiz Belardinelli[12] on behalf of the INSTANT Investigators

[1]MUMC, Maastricht, NL; [2]Isala Clinics, Zwolle, NL; [3]Deventer Hospital, Deventer, NL; [4]Spaarne Gasthuis, Haarlem, NL; [5]OLVG, Amsterdam, NL; [6]Long Beach Memorial Medical Center, CA, USA; [7]Slingeland Ziekenhuis, Doetinchem, NL; [8]University of Groningen, Groningen, NL; [9]St. George's University, London, UK [10]Mass General, MA, USA; [11]Lankenau Heart Institute, PA, USA; [12]InCarda Therapeutics, CA, USA.

Background

- Atrial fibrillation (AF) is the most common cardiac arrhythmia with an estimated (2010) global prevalence of 33.5 million.[1] Paroxysmal AF represents approximately 30% of all AF cases.
- In patients with AF, sinus rhythm (SR) can be re-established by either electrical cardioversion (ECV) or pharmacological cardioversion (PCV). In Europe, there are several options for acute PCV (e.g., IV flecainide, IV propafenone, IV vernakalant, IV ibutilide); whereas in the US there are fewer options.
- In the US, IV ibutilide is the only antiarrhythmic agent currently approved for acute PCV of AF. Ibutilide has a conversion rate of AF to SR of ~40% in patients with recent-onset AF; however, its use is limited due to the risk of Torsades de Pointes and the requirement for at least 4 hours of in-hospital monitoring.[2,3]
- In Europe, IV flecainide is a first-line therapy for pharmacological cardioversion of AF in patients who are hemodynamically stable and have minimal or no structural heart disease.[4]
- Clinically important adverse events (AEs) known to be associated with acute PCV of recent-onset AF with antiarrhythmic agents (including IV flecainide) are bradycardia, hypotension, atrial flutter (AFL) with rapid ventricular rate (i.e., AFL with 1:1 AV nodal conduction), ventricular tachycardia (VT), and post-cardioversion sinus pause (and/or ventricular pause). These AEs are herein referred to as cardiovascular (CV) events of special interest.[5,6,7]
- The INSTANT (INhalation of flecainide to convert recent-onset SympTomatic Atrial fibrillatioN to sinus rhyThm) trial was an open-label, multicenter study of flecainide acetate oral inhalation solution (FlecIH) for acute conversion of recent-onset, symptomatic atrial fibrillation (AF) to sinus rhythm (SR).[8]
- The initial dose-ranging study (Part A) established safety and feasibility, while showing a dose- and concentration-dependent increase in efficacy (dose range: 30-120 mg). The highest dose (120 mg) was selected for further evaluation in a second study (Part B).

InRhythm (Orally Inhaled Flecainide)

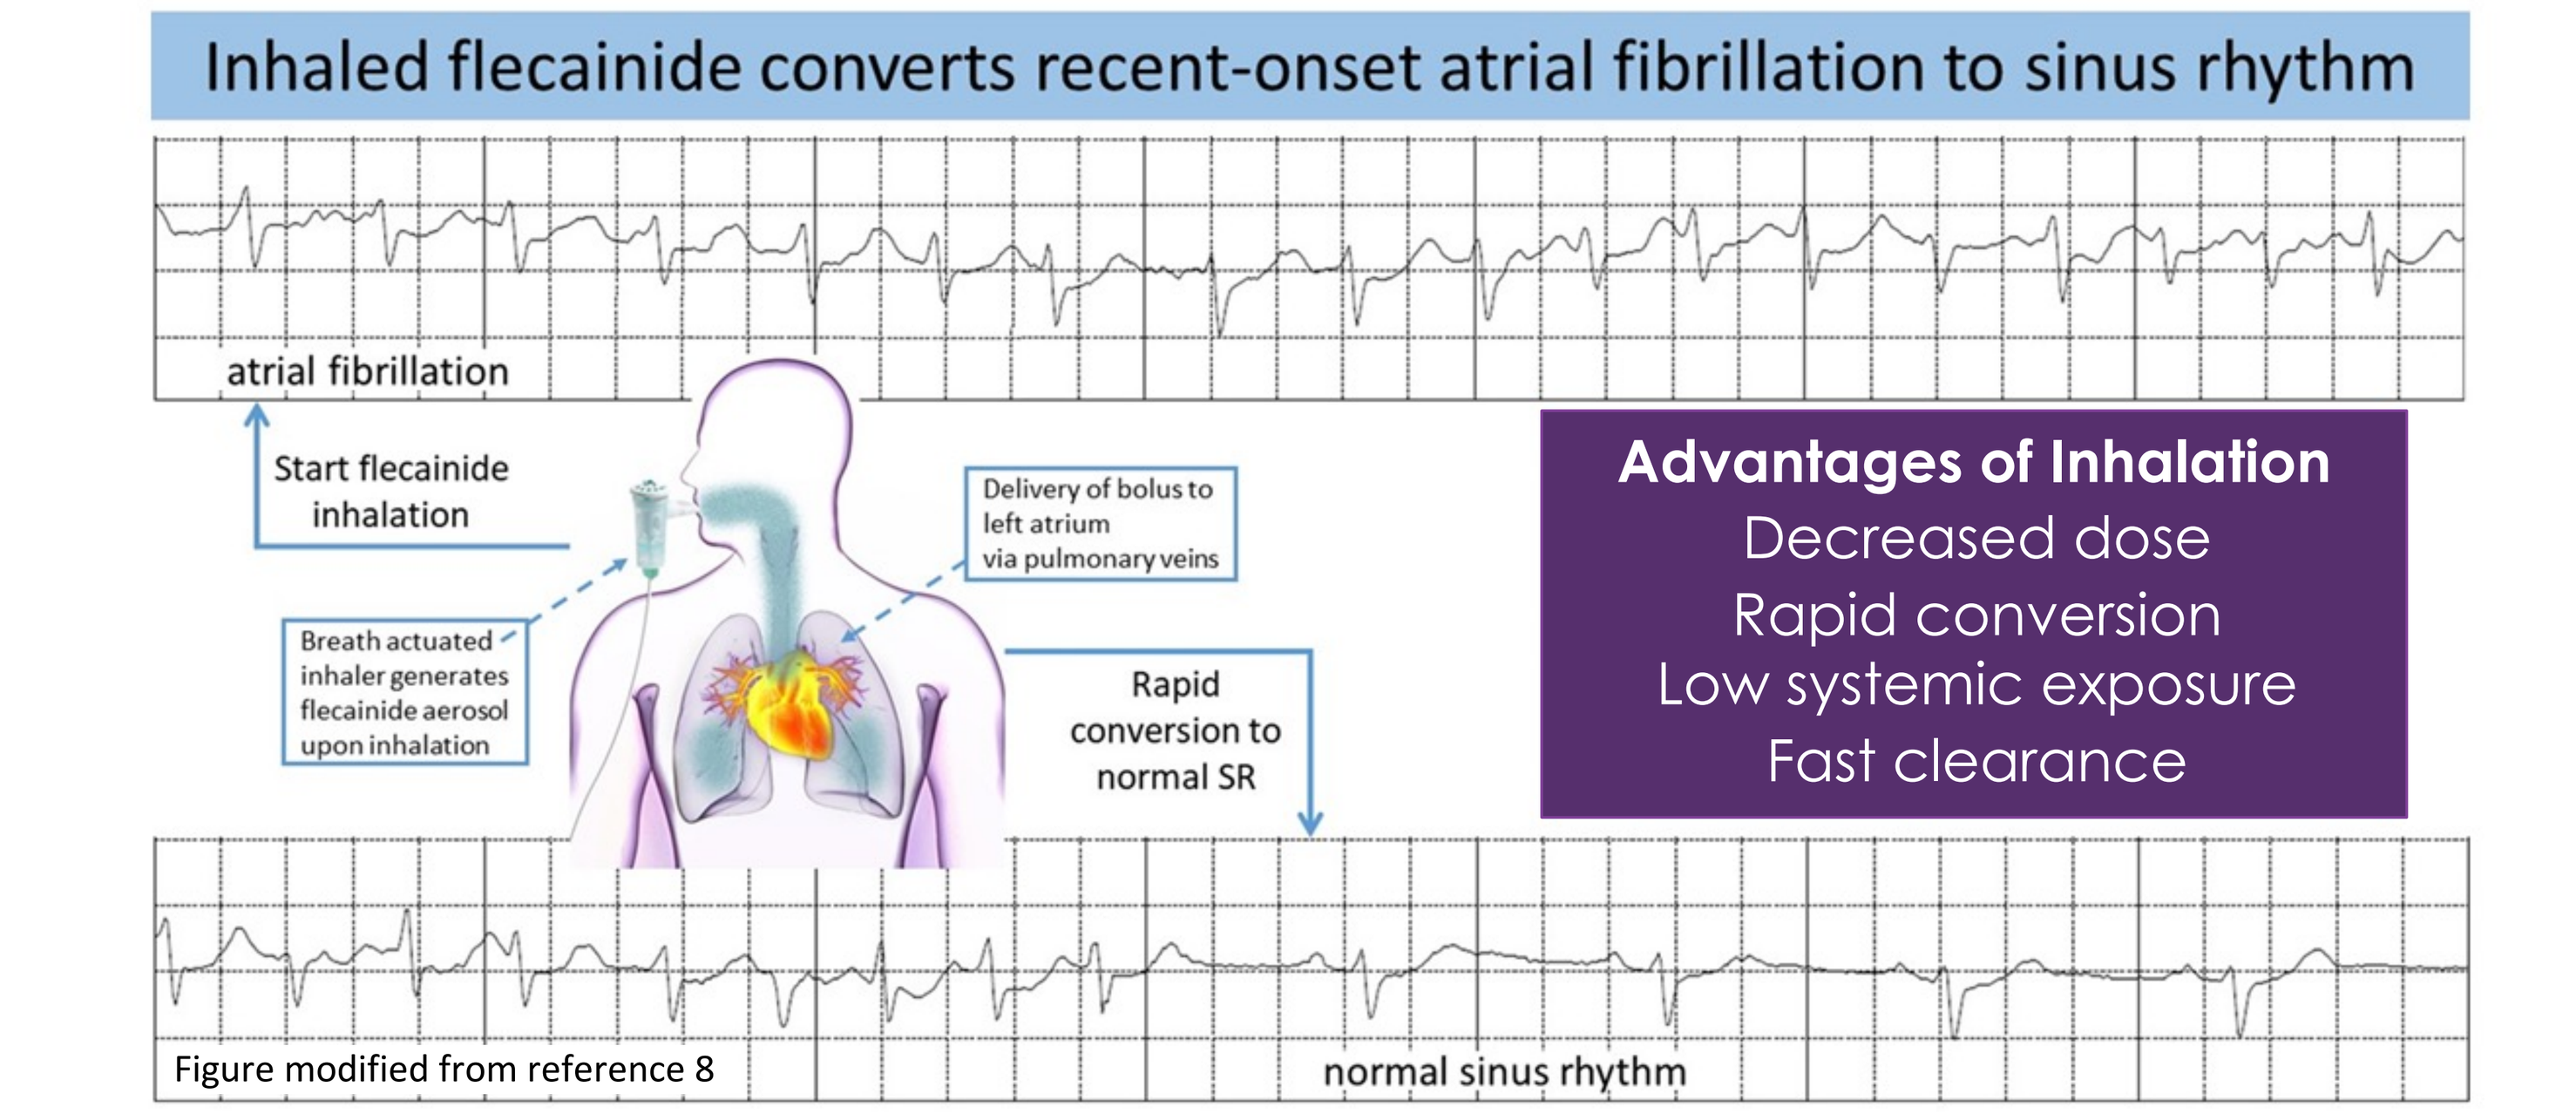
Inhaled flecainide converts recent-onset atrial fibrillation to sinus rhythm
atrial fibrillation
Start flecainide inhalation
Breath actuated inhaler generates flecainide aerosol upon inhalation
Delivery of bolus to left atrium via pulmonary veins
Rapid conversion to normal SR
Advantages of Inhalation
Decreased dose
Rapid conversion
Low systemic exposure
Fast clearance
Figure modified from reference 8
normal sinus rhythm

Figure 1: InRhythm (orally inhaled flecainide) for conversion of recent onset atrial fibrillation

- Oral inhalation of 120 mg eTLD (estimated total lung dose) is self-administered over an 8-minute period (two 3.5-minute inhalation periods separated by a 1-minute break) using a hand-held, breath-actuated nebulizer (AeroEclipse II BAN).

Methods

- Patients were monitored using 12-lead electrocardiogram and a 4-hour Holter; vital signs and adverse events (AEs) were recorded. Patients who did not convert to SR were offered alternative treatment per the investigator's discretion.
- Data are presented for the safety populations in the Part A (N=29) and Part B (N=25) who received 120 mg FlecIH-103, and for the combined Part A/B cohort (N=54).

Patient Baseline Characteristics

Table 1: Baseline Characteristics by Study Cohort

Characteristic	Part A (N = 29)	Part B (N = 25)	All Patients (N=54)
Age (y)	62.6 ± 12.4	61.7 ± 11.0	62.1 ± 11.7
Male sex, n (%)	19 (65.5%)	17 (68.0%)	36 (66.7%)
White, n (%)	26 (89.7%)	24 (96.0%)	50 (92.6%)
Body mass index (kg/m2)	26.7 ± 3.7	26.9 ± 3.8	26.8 ± 3.8
Hypertension, n (%)	9 (31.0%)	7 (28.0%)	16 (29.6%)
Hyperlipidemia, n (%)	9 (31.0%)	6 (24.0%)	15 (27.8%)
Diabetes, n (%)	1 (3.4%)	0 (0.0%)	1 (1.9%)
NYHA HF Class I, n (%)	3 (10.3%)	3 (12.0%)	6 (11.1%)
NYHA HF Class II, n (%)	1 (3.4%)	0 (0.0%)	1 (1.9%)
CHA_2DS_2-VASc Score	1.6 ± 1.5	1.1 ± 1.0	1.4 ± 1.3
AF Sx Duration ≥ 1 and ≤ 24 hours, n (%)	25 (86.2%)	20 (80.0%)	45 (83.3%)
AF Sx Duration ≥ 24 and ≤ 48 hours, n (%)	4 (13.8%)	5 (20.0%)	9 (16.7%)
First AF Episode, n (%)	13 (44.8%)	4 (16.0%)	17 (31.5%)
Recurrent Paroxysmal AF Episode, n (%)	15 (51.7%)	18 (72.0%)	33 (61.1%)
AF Post-Cardiac Ablation, n (%)	1 (3.4%)	3 (12.0%)	4 (7.4%)
# Previous AF Episodes (excludes 1st episode pts)	3.1 ± 3.7	2.7 ± 3.3	2.8 ± 3.4

Data are mean ±SD unless otherwise noted.
Safety population (N=54)

- Baseline characteristics were similar in the two cohorts; however, the Part A cohort had a higher proportion of patients treated for their first AF episode compared to the Part B cohort that included more patients with a recurrent paroxysmal AF episode.

Pharmacokinetics & Pharmacodynamics

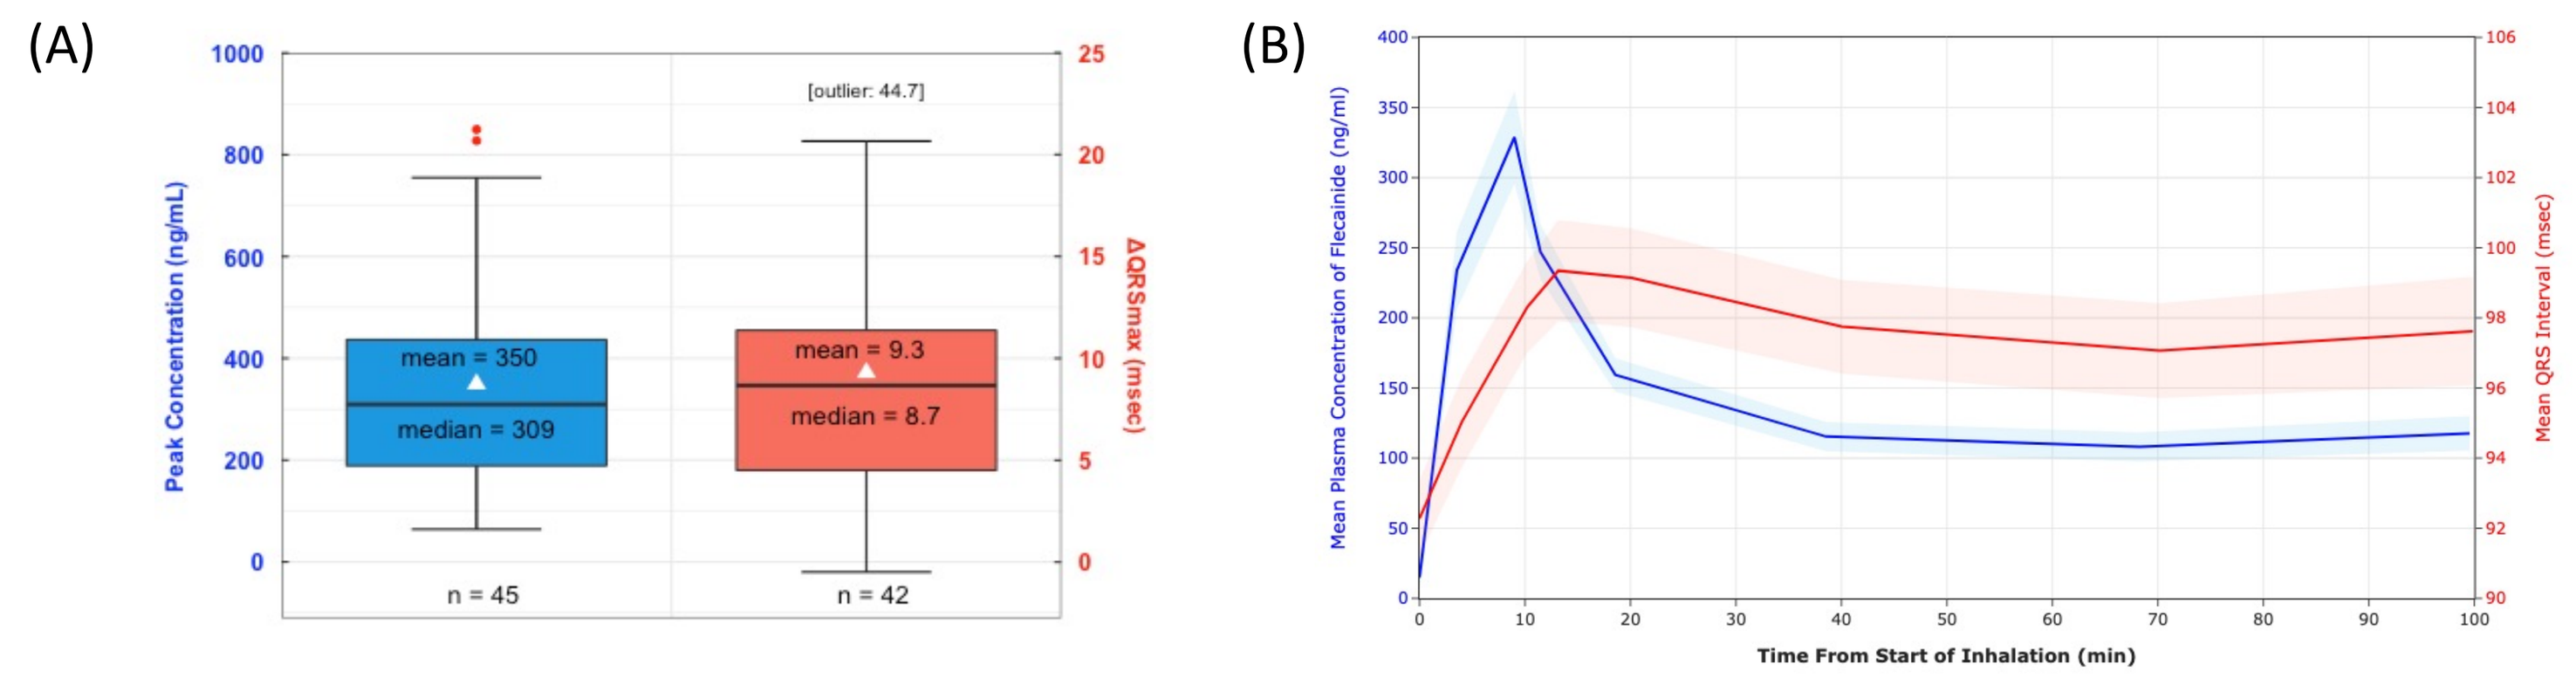
(A)
Peak Concentration (ng/mL)
ΔQRSmax (msec)
[outlier: 44.7]
mean = 350
median = 309
mean = 9.3
median = 8.7
n = 45
n = 42
(B)
Mean Plasma Concentration of Flecainide (ng/ml)
Mean QRS Interval (msec)
Time From Start of Inhalation (min)

Figure 2: (A) Maximum plasma concentration of flecainide and (B) QRS interval (PK population; N=45/42)

- Flecainide peak plasma concentration (mean±SD) was similar in Part A (373±189 ng/mL), Part B (323±218 ng/mL), and in the combined cohort (350±202 ng/mL); no patient had a peak plasma concentration of flecainide ≥ 1000 ng/mL.
- ΔQRSmax (mean±SD) was similar in the Part A (8.2±4.7 msec), Part B cohort (8.7±8.9 msec), and in the combined cohort (8.4±6.8 msec).
- One patient had a ΔQRSmax >30msec. This patient had a baseline QRS of 103 msec and had rate dependent right bundle branch block (RBBB). At 2 minutes after completion of inhalation a transient RBBB was observed. Vital signs were stable throughout the 90-minutes observation period.

Time to Conversion of AF to SR

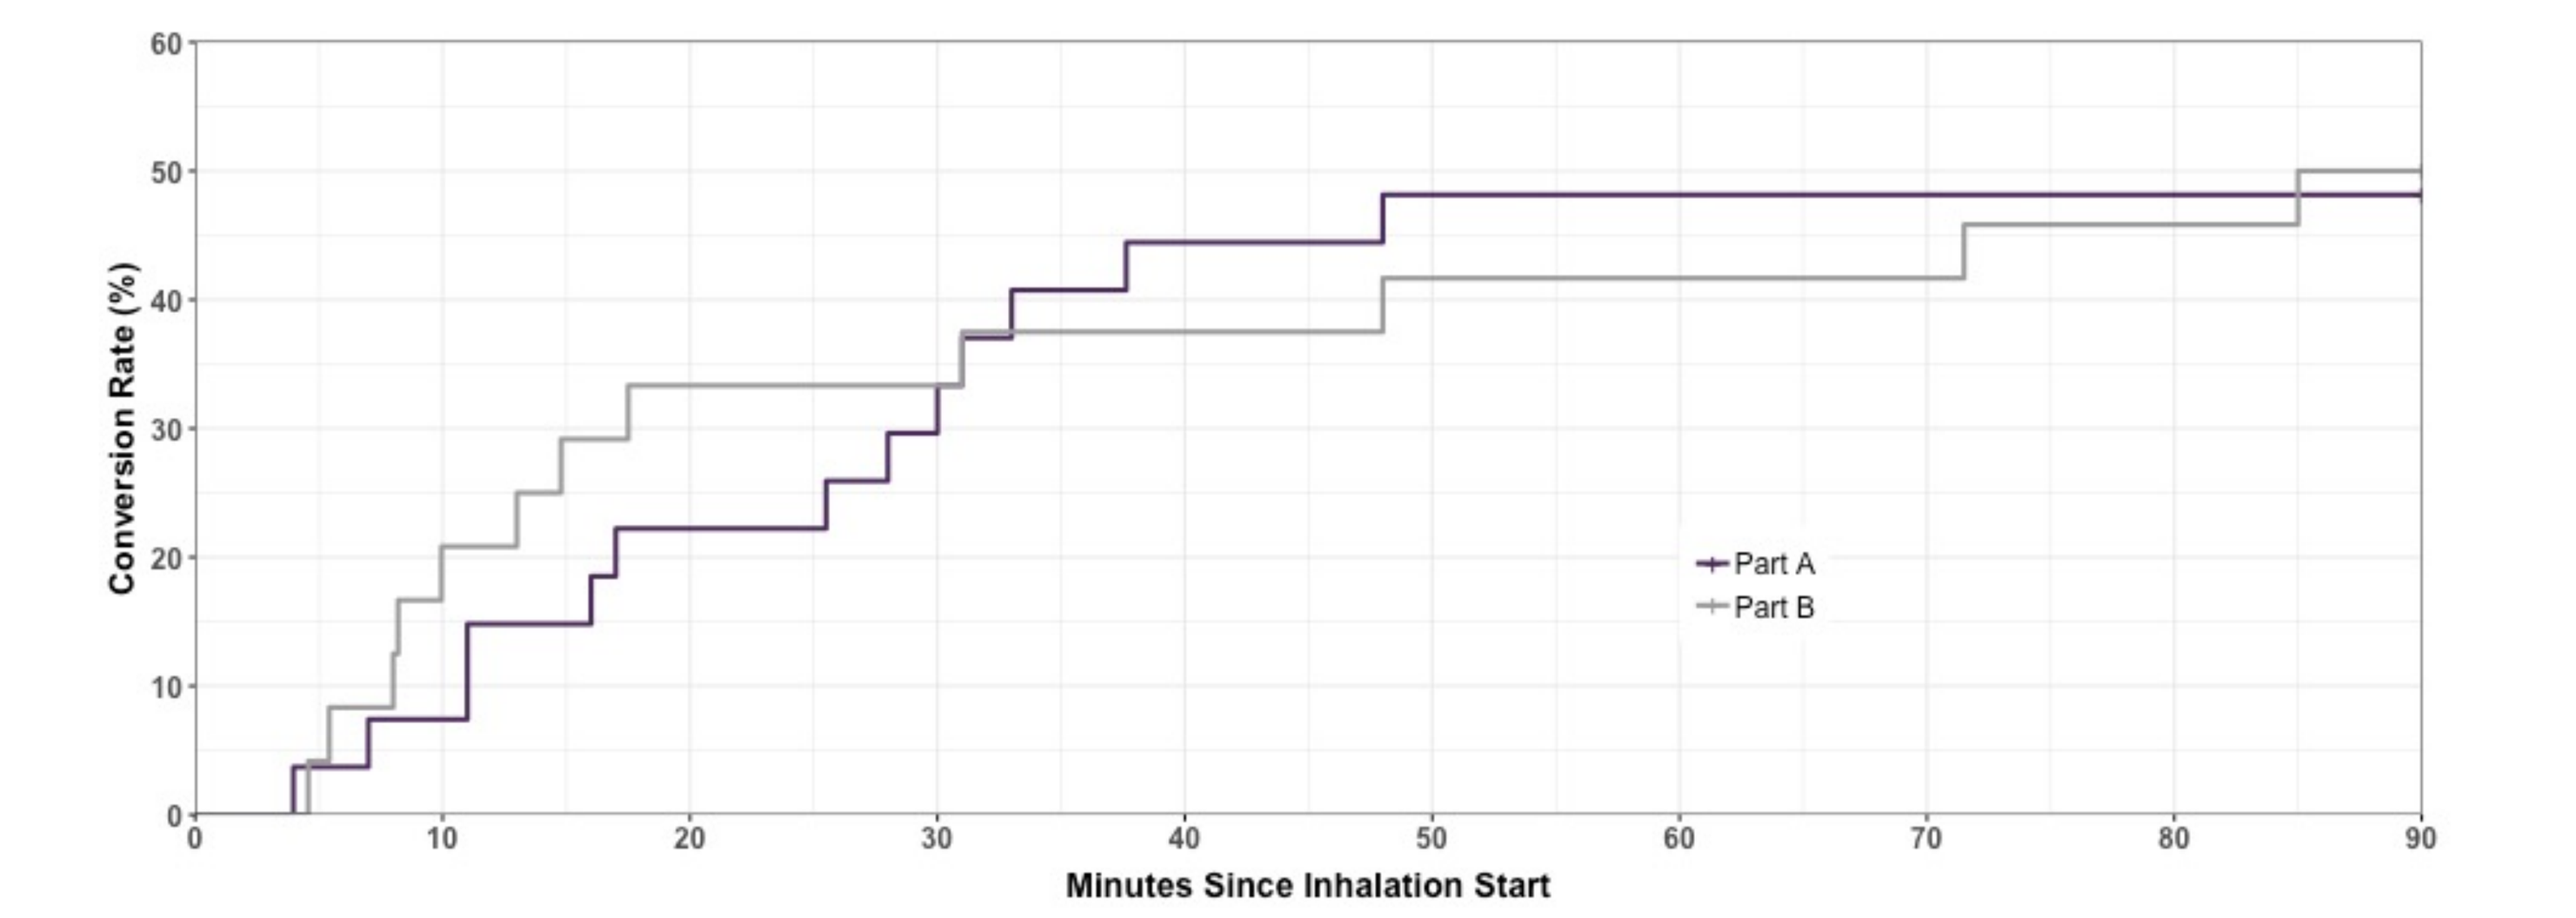
Conversion Rate (%)
Minutes Since Inhalation Start
Part A
Part B

Figure 3: Time to conversion of AF to SR with inhaled flecainide (mITT population; N=27/24)

- The conversion rate at 90 minutes post-dose was 48% (95% CI: 28.7%, 68.1%) in Part A, 50% (95% CI: 29.1%, 70.9%) in Part B, and 49% (95% CI: 34.8%, 63.4%) for the combined cohort.
- Time to conversion was similar in the two cohorts, with a median time of 6.9 minutes (IQR: 1.2, 21.6 minutes) from the end of the inhalation in the combined cohort.

Adverse Events

Table 2: Frequency of Treatment Emergent Adverse Event by Preferred Term

Preferred Term	Part A (N = 29)	Part B (N = 25)	All Patients (N=54)
Cough*	12 (41.4%)	11 (44.0%)	23 (42.6%)
Dyspnea	3 (10.3%)	4 (16.0%)	7 (13.0%)
Oropharyngeal pain	4 (13.8%)	3 (12.0%)	7 (13.0%)

Number (%) of subjects with at least 1 AE for all preferred terms reported in >10% in the combined cohort.
*Epoch of reflex cough lasting several seconds only.

- The most frequently reported AEs were cough, dyspnea, and oropharyngeal pain; these AEs were transient, and none led to discontinuation of inhalation.
- Two patients experienced CV events of special interest that were considered serious (bradycardia and atrial flutter with 1:1 AV conduction) but neither patient required treatment and both events resolved without sequelae.

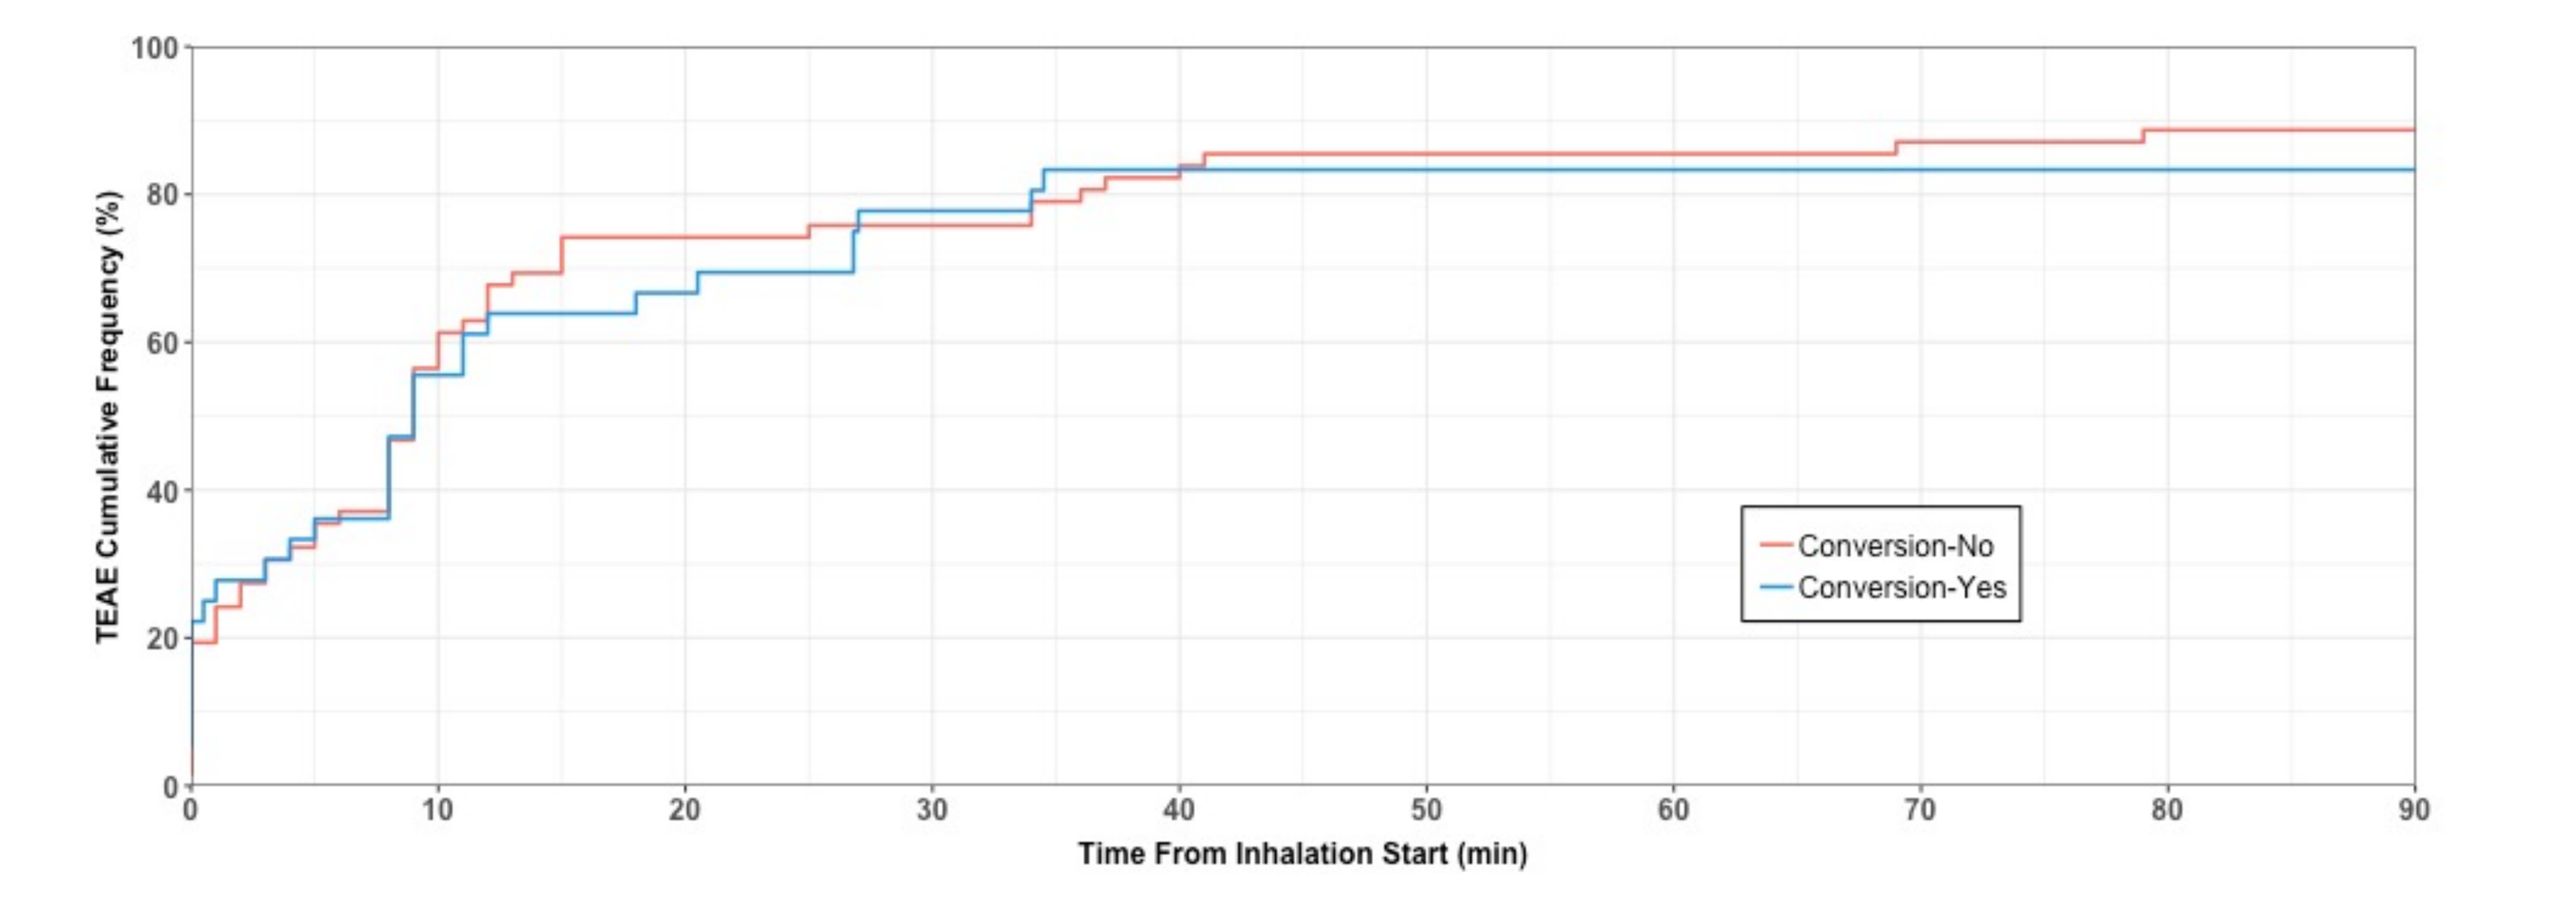
TEAE Cumulative Frequency (%)
Time From Inhalation Start (min)
Conversion-No
Conversion-Yes

Figure 4: Time of onset for treatment emergent AEs by conversion status (safety population; N=54)

- Nearly all (>80%) treatment emergent adverse events (TEAEs) occurred within 40 minutes of the start of dosing and were of limited duration.
- The onset of TEAEs was similar in patients whose AF converted to SR compared to patients whose AF did not convert to SR with inhaled flecainide.

AF Symptoms

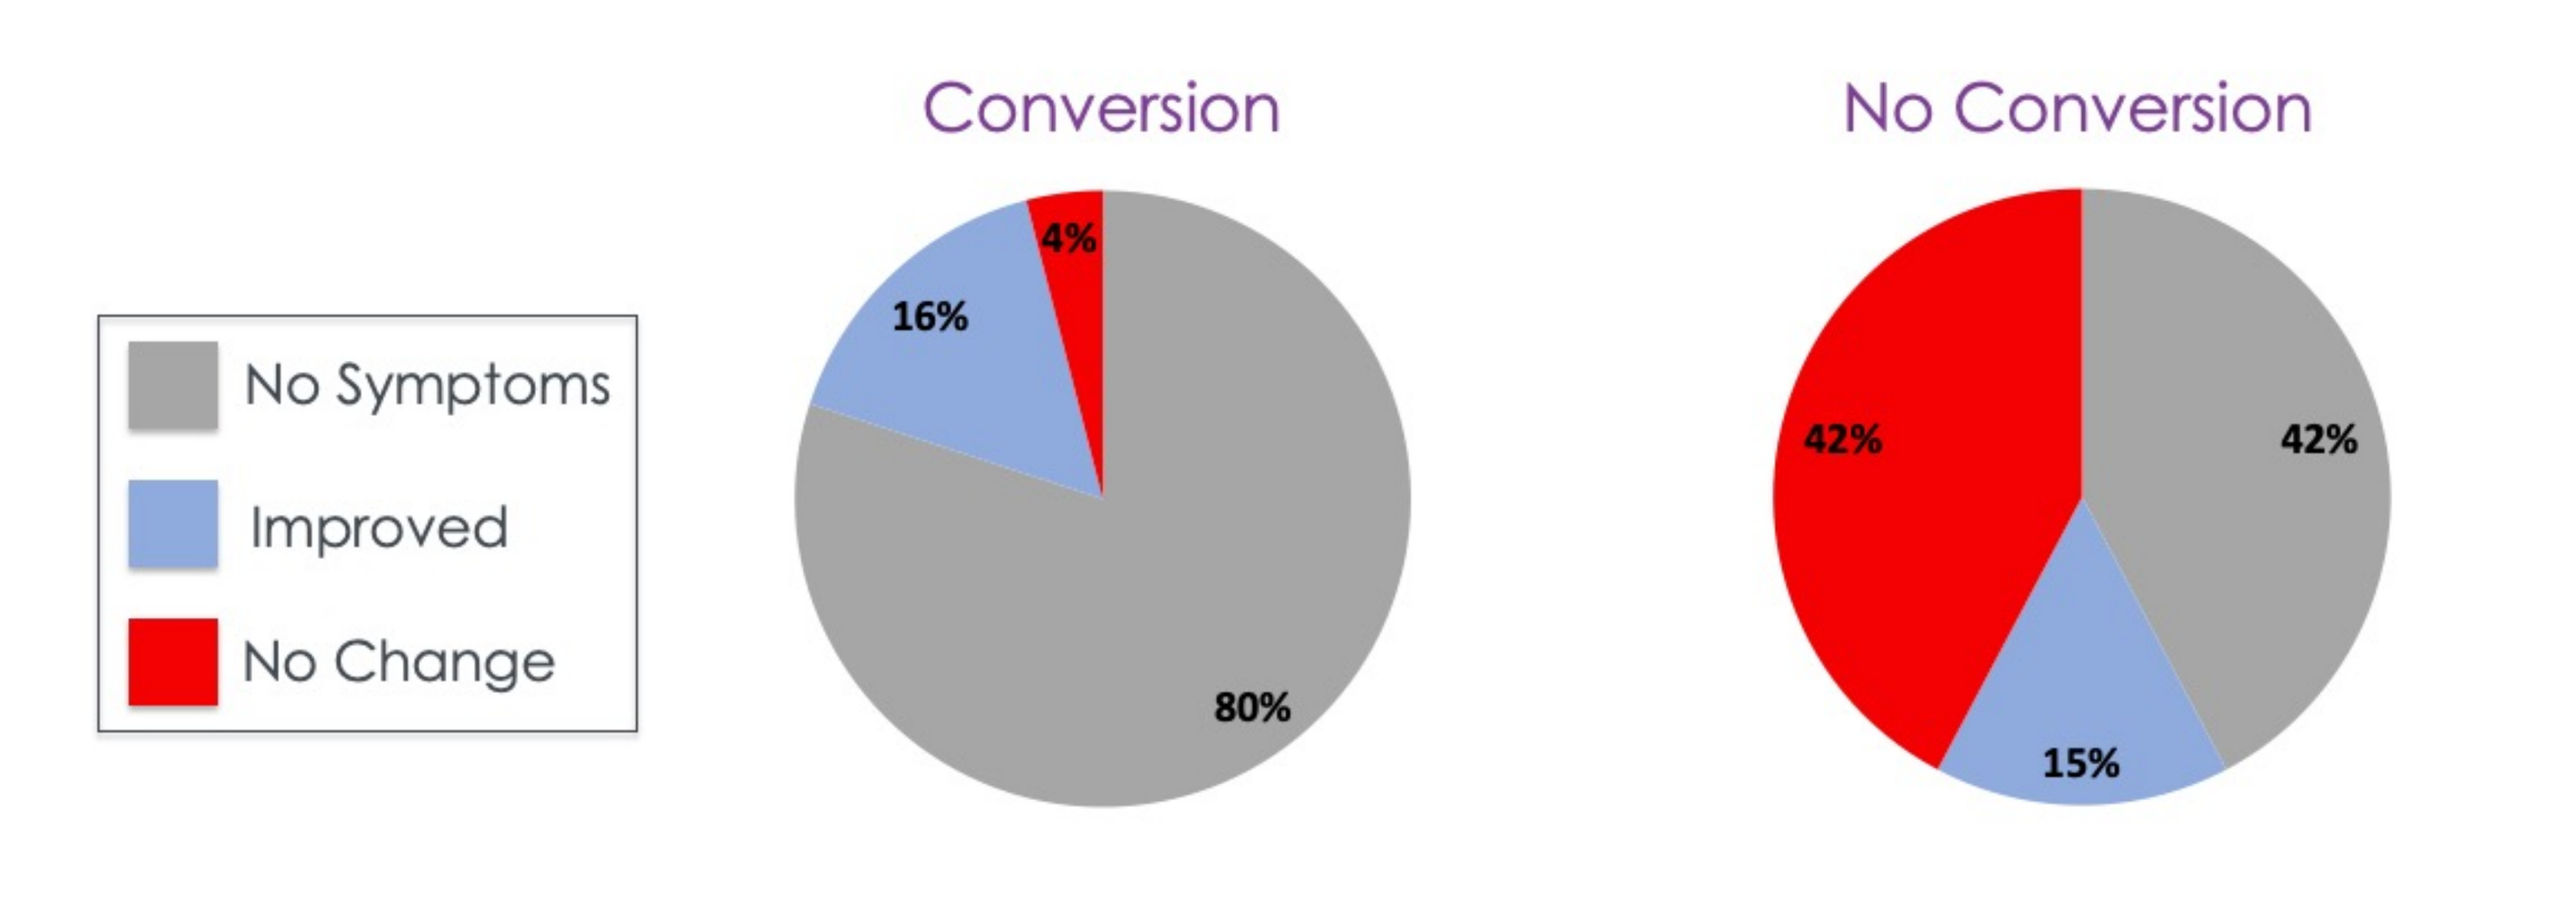
Conversion
No Conversion
No Symptoms
Improved
No Change
4%
16%
80%
42%
42%
15%

Figure 5: AF symptoms at 90 minutes post-dose by conversion status (mITT population; N=51)

- At baseline, all patients (100%) had at least one AF-related symptom: palpitations (84%), chest discomfort (41%), dizziness (37%), shortness of breath (35%)
- A greater proportion of patients whose AF converted to SR with inhaled flecainide reported no symptoms or improved symptoms at 90 minutes compared to patients whose AF did not convert to SR (p = 0.002).

Conclusions

- The conversion rate of recent onset AF to SR following 120 mg flecainide administered via oral inhalation is similar to that reported for oral and IV flecainide.
- No patients had a flecainide peak plasma level >1000 ng/ml, a concentration that is associated with significant increase in the risk of serious CV events.
- The mean maximum QRS prolongation was ∽3-fold lower than that reported for IV flecainide.
- Nearly all adverse events and all serious CV events of special interest had an onset of less than 40 minutes from the start of inhalation.
- The risk-benefit of orally inhaled flecainide acetate inhalation solution for acute cardioversion of recent onset AF is highly favorable and may provide a safe, effective, and more convenient therapeutic option compared to either ECV or PCV with IV antiarrhythmic drugs.
- A 400-patient, placebo-controlled Phase 3 trial (RESTORE-1) will begin enrollment in the United States, Canada, and Europe in the first half of 2022.

References

1. Chugh SS, Havmoeller R, Narayanan K, et al. (2014). Circulation, 129: 837-847.
2. Ellenbogen KA, Stambler BS, Wood MA et al. (1996). JACC 28(1): 130-136.
3. Corvert (ibutilide fumarate) US Package Insert (2001).
4. Hindricks G, Potpara T, Dagres N, et al. (2021). Eur Heart J, 42:373-498.
5. Aliot E, Capucci A, Crijns H, et al. (2011). Europace, 13: 161-173.
6. Tamargo J, Capucci A and Mabo P (2012). Drug Saf, 35(4): 273-289.
7. Markey GC, Salter N, Ryan J (2018). J Emerg Med, 54(3):320-327.
8. Crijns HJ, Elvan A, Al-Windy N, et al. (2022). Circ Arrhythm Electrophysiol, 15(3): 157-164.

InCarda Therapeutics, Inc.
Maastricht UMC+